CAN-FITE BIOPHARMA, LTD.
10 Bareket Street, Kiryat Matalon, P.O.Box 7537
Petach Tikva 49170, ISRAEL
Tel: +972-3-9241114
Fax: +972-3-9249378
E-mail:sara@canfite.co.il
Web site: www.canfite.com

Poster no. 384

Phase 1/2 Trial of CF102, a Selective A_3 Adenosine Receptor (A_3AR) Agonist, in Patients with Hepatocellular Carcinoma

[1]SM Stemmer, [1]O Benjaminov, [1]G Medalia, [2]MH Silverman, [2]WD Kerns, [2]S Bar-Yehuda, [2]S Fishman, [2]Z Harpaz, [2]M Farbstein, [2]P Fishman

[1]Institute of Oncology, Davidoff Center, Rabin Medical Center, Sackler School of Medicine, Tel Aviv University, Petach-Tikva, Israel

[2] Can-Fite BioPharma Ltd., Kiryat-Matalon, Petach-Tikva, Israel

CF102

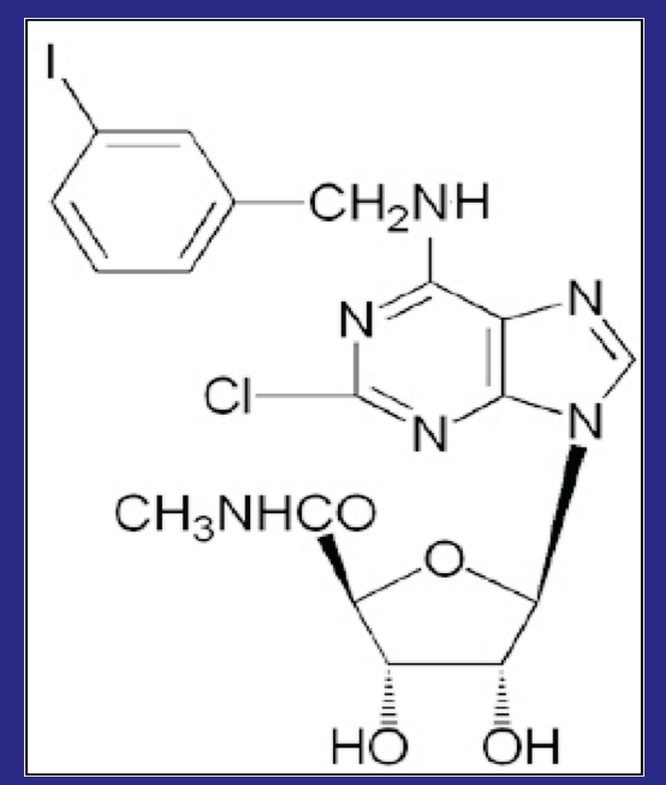
I
CH_2NH
N
N
Cl
N
N
CH_3NHCO
O
HO
OH

2-chloro-N[6]-(3-iodobenzyl)-adenosine-5'-N-methyl-uronamide, also known as Cl-IB-MECA

Background

The Gi protein associated cell surface receptor, A_3 adenosine receptor (A_3AR) was suggested as a target for cancer treatment. The A_3AR agonist 2-chloro-N^6-(3-iodobenzyl)-adenosine-5'- N-methyl-uronamide (CF102), was found to inhibit the growth of hepatocellular carcinoma in a syngeneic and xenograft experimental animal models (Figure 1).[1]

Figure 1. CF102 inhibits hepatocellular carcinoma growth in animal model

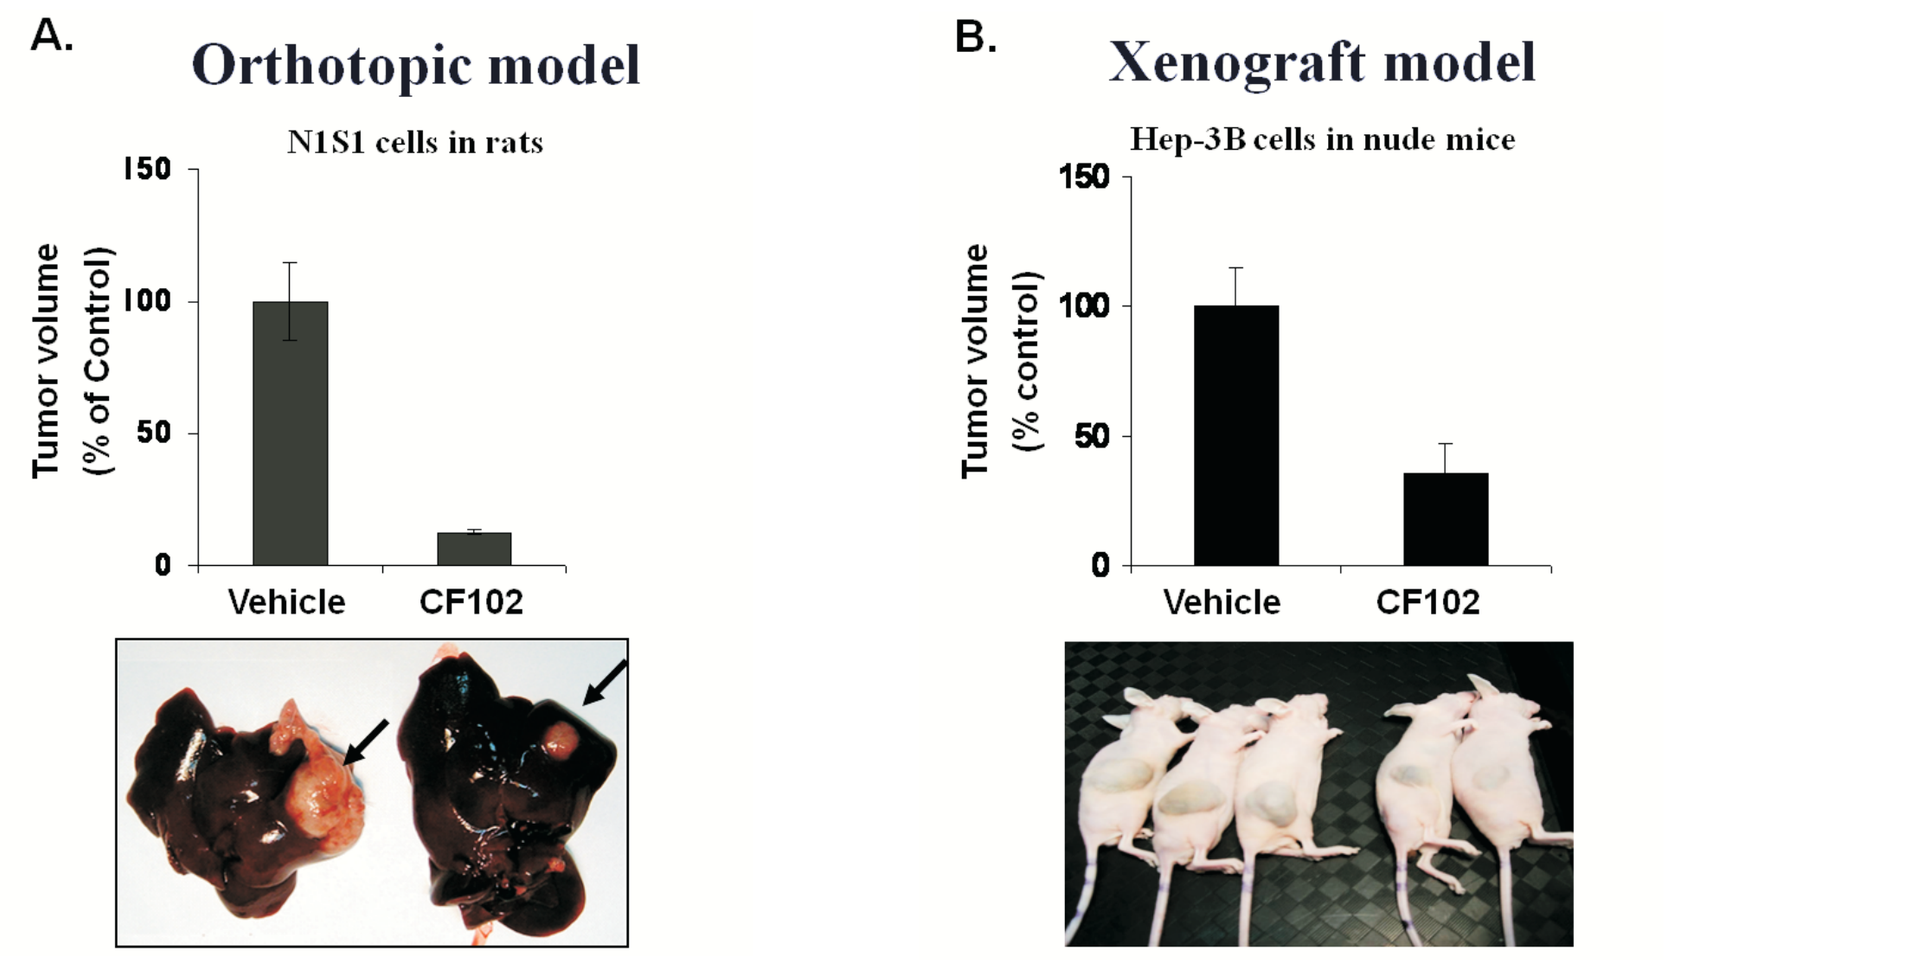
A.
Orthotopic model
N1S1 cells in rats
Tumor volume (% of Control)
150
100
50
0
Vehicle
CF102
B.
Xenograft model
Hep-3B cells in nude mice
Tumor volume (% control)
150
100
50
0
Vehicle
CF102

A. In the syngeneic orthotopic model, N1S1 cells were inoculated into the liver of rats and oral treatment with CF102 (100µg/kg) was initiated 3 days after tumor inoculation. CF102 treatment exerted a marked inhibitory effect (92.8±6.9%) on tumor growth.

B. In the Hep-3B xenograft model, cells ($2.5x10^6$) were subcutaneously injected into the flank of balb/c nude mice. When the tumor reached ~50-100mm^3 in size, CF102 (100µg/kg) was orally administered. CF102 treatment resulted in a significant inhibition of tumor growth.

Mechanistically, CF102 induces de-regulation of the NF-κB and the Wnt signal transduction pathways, resulting in apoptosis of the hepatocellular carcinoma cells (Scheme 1a)[1].

Furthermore, CF102 exerts an anti-viral effect, demonstrated by its ability to inhibit the replication of Hepatitis C virus, utilizing Replicon assay and infectious virus cell culture assay in hepatoma cells. In the latter, the anti-viral effect was mediated via the down-regulation of the non-structural 5A protein (NS5A) expression levels (Scheme 1b).

CF102 also induces an anti-inflammatory effect, mediated via down-regulation of the NF-κB, in an experimental animal model of acute hepatitis (Scheme 1c).

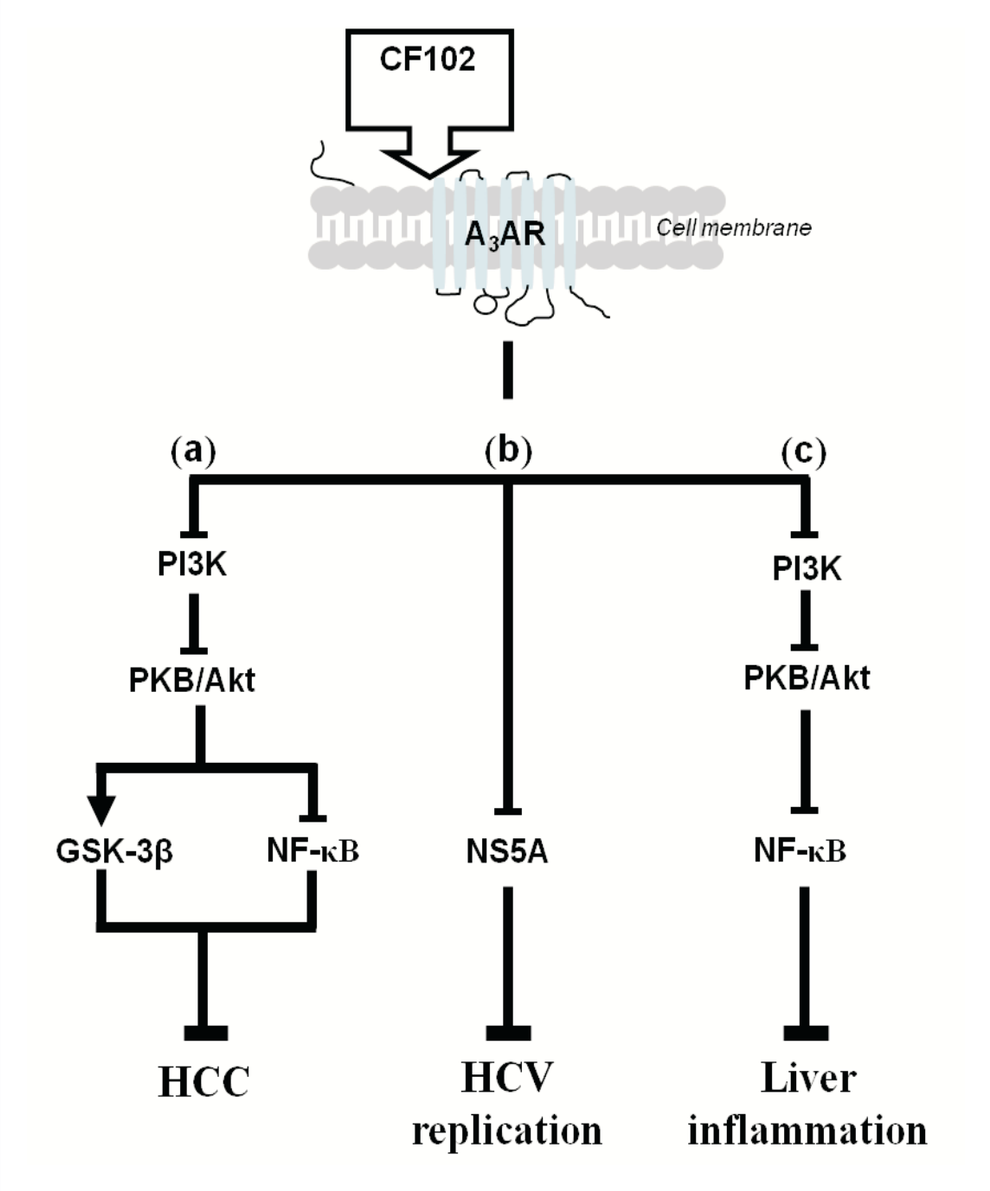
CF102
A_3AR
Cell membrane
(a)
(b)
(c)
PI3K
PKB/Akt
GSK-3β
NF-κB
NS5A
PI3K
PKB/Akt
NF-κB
HCC
HCV replication
Liver inflammation

Pre-clinical and Phase I studies demonstrated that CF102 is safe and well tolerated.

The current Phase 1/2 study is aimed to look at the safety and efficacy of CF102 in patients with hepatocellular carcinoma. The study continues to recruit patients.

[1]Bar Yehuda, Int. J. Oncology, 2008

Study Objectives

- To determine the safety and tolerability, dose-limiting toxicities (DLTs), maximum tolerated dose (MTD), and recommended Phase 2 dose (RP2D) of orally administered CF102 in patients with advanced hepatocellular carcinoma (HCC)
- To assess the repeat-dose pharmacokinetic (PK) behavior of CF102 in patients with HCC
- To document any observed therapeutic effect of CF102 in patients with HCC

Methods

Patients

Adults diagnosed with HCC (by cytology and/or histology or according to the American Association for the Study of Liver Diseases Practice Guideline algorithm) with or without underlying cirrhosis enrolled to the study. HCC is advanced, refractory, or metastatic, and no standard therapies are expected to be curative. The conduct of this trial was approved by all local Ethics Committees in accordance with the Declaration of Helsinki. Written informed consent was obtained from the patients before inclusion in the study.

Study design

This was a Phase 1/2, open-label, dose-escalation study. Utilizing a "3+3" design, the initial dose of oral CF102 in the 3 patients in Cohort 1 was 1 mg BID, with subsequent escalations up to 5 and 25 in continuous cycles of 28 days each unless limited by toxicity.

Safety was assessed according to of Dose-Limiting Toxicity); characterization of the type, incidence, severity (graded by National Cancer Institute Common Terminology Criteria for Adverse Events), timing, seriousness, and relationship to treatment of adverse events; effects on vital signs and laboratory parameters; and changes in electrocardiograms (ECGs).

Pharmacokinetic was evaluated by PK parameters, including area under the curve (AUC), maximum plasma concentration (Cmax), trough plasma concentration (Cmin), time to maximum plasma concentration (Tmax), and plasma halflife (t1/2) will be determined. Comparisons across dose levels will be made to assess proportionality. In addition, comparison between single-dose and multi-dose PK parameters will be made for assessment of steady-state drug accumulation.

Patients with good performance status were allowed at progression to be escalated to the next higher cohort dose

Current Data From the Ongoing Study

Patients

Overall 11 patients, median age (64-90) years, Child-Pugh Class A or B, were treated with CF102. The first 9 patients were enrolled across 3 cohorts, 3 patients at each dose level 1, 5, 25 mg. For efficacy dose finding, 3 patients are currently enrolled at each dose level.

Table 1: Patients' baseline characteristics and disposition

Patient	CF102 Dose	Gender	Age (years)	Disease duration (years)	Previous medications
1	1mg BID	Male	80	5.8	DOXORUBICIN; RADIATION
2	1mg BID	Male	90	1.5	
3	1mg BID	Female	72	1.3	
4	5mg BID	Female	78	2.3	SORAFENIB; RESECTION OF LIVER MASS
5	5mg BID	Male	64	2	DOXORUBICIN; SORAFENIB RADIATION TO SPENAODAL SINUS METASTASIS
6	5mg BID	Male	74	2.6	DOXORUBICIN; SORAFENIB +/- ERLOTINIB
7	25mg BID	Female	76	1.8	SORAFENIB+/- ERLOTINIB
8	25mg BID	Female	86	0.58	
9	25mg BID	Male	69	0.58	
10	5mg BID	Female	73	1	
11	25mg BID	Male	71	1.3	SORAFENIB; TACE

Safety

- At any dose level no dose-limiting toxicities, specifically attributed to CF102, were observed
- All events classified as drug-related were either grade 1 or 2
- No drug-related abnormalities of hematologic, renal, or hepatic function have been observed on laboratory testing

Table 2: Adverse events

Event	No. of patinets
Anorexia/loss of appetite	5
Abdominal pain	4
Asthenia/weakness	4
Diarrhea	3
Leg edema/swelling	2
Fatigue	2
Fever	2
Nausea	2
Back pain	2
Chest pain	2
Leg pain	2

Table 3: Serious adverse events

	Event	Drug relationship
Patient 1	Inflammation of the knee	Not related
	Cellulitis	Not related
	Cirrhosis	Not related
	Swelling of legs	Not related
	General deterioration	Not related
Patient 2	Disease progression	Not related
Patient 3	Cerebral hemorrhage	Possibly related
	Headache	Possibly related
	Hypercalcemia	Not related
	Hyponatermia	Not related
	Oliguria	Not related
	Asystole	Not related
Patient 4	Leg swelling due to superficial and deep vein thombosis	Possibly realted

Pharmacokinetics

CF102 has shown good oral bioavailability and linear PK behavior given at single (Figure 2A) or repeated doses (Figure 2B). Plasma levels at all doses are above the affinity value of CF102 to the A_3AR.

Figure 2A. Pharmacokinetic behavior of CF102 after single dose — day 1 of Cycle 1

Figure 2B. Pharmacokinetic behavior of CF102 after repeated dose at steady state — day 29 of Cycle 1

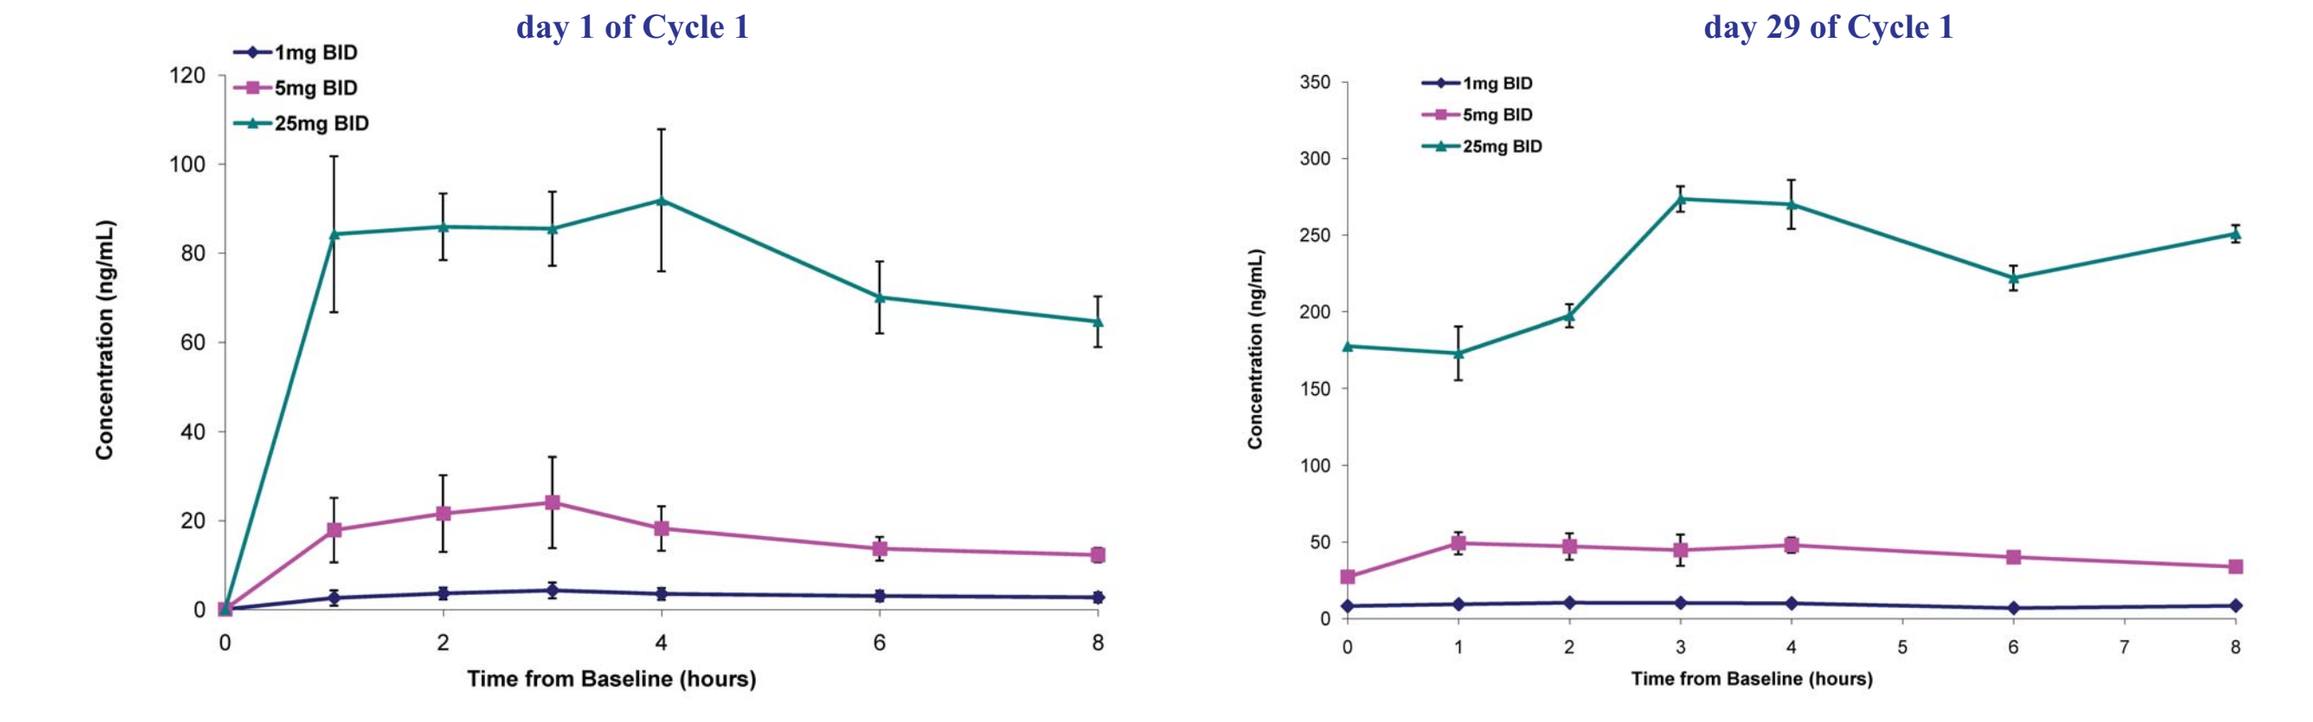
1mg BID
5mg BID
25mg BID
Concentration (ng/mL)
Time from Baseline (hours)

Clinical Findings

- **Survival** - Currently, 7 out of first 9 patients are under CF102 treatment for 6-13 months
- **Regression of skin metastasis** - Complete clinical regression of biopsy-proven skin metastases was observed in one patient (Figure 3)
- **Hepatitis C inhibition by CF102** - CF102 decreased viral load in 3 patients with liver cancer who were also infected with hepatitis C virus with no rebound after 6 months in one of them (Figure 4)

Figure 3. Cytology of skin metastases before treament with CF102

Malignant cells having large hyperchromatic nuclei with nucleo-cytoplasmatic ratio>1

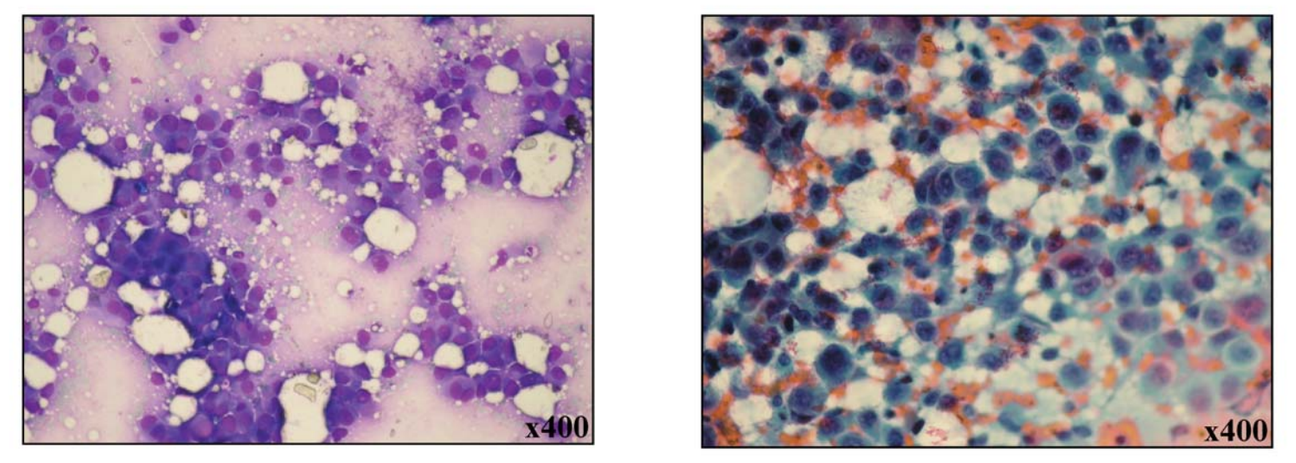
x400
x400

GIMSA staining — PAPANICOLAU staining

Figure 4. CF102 reduced HCV viral load in HCC patients infected with HCV

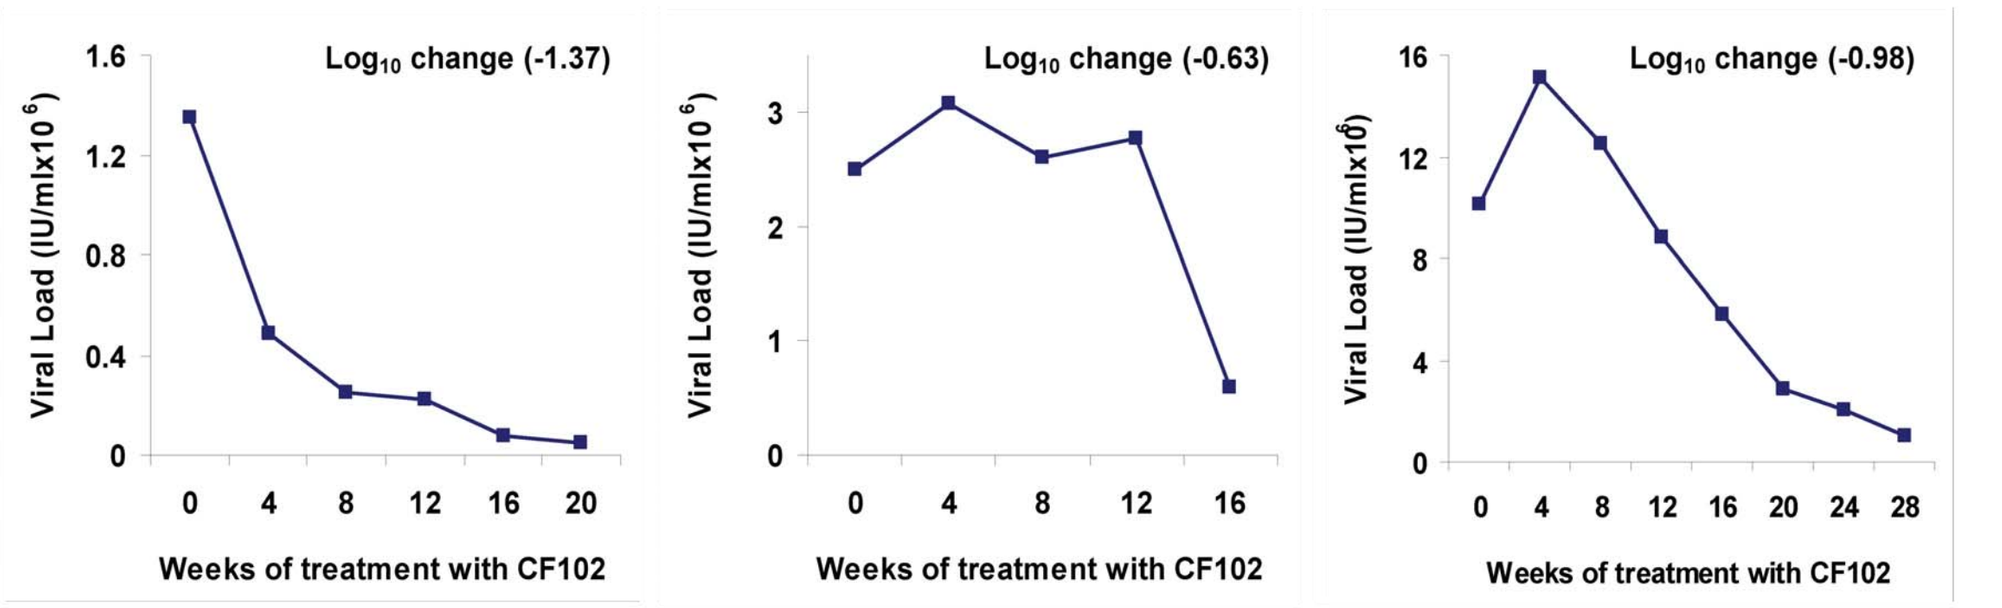
Log10 change (-1.37)
Log10 change (-0.63)
Log10 change (-0.98)
Viral Load (IU/mlx10⁶)
Weeks of treatment with CF102

Conclusions

- Daily oral CF102 is safe and well tolerated at doses up to 25 mg twice daily and shows linear PK in patients with HCC
- CF102 has shown preliminary evidence of clinical activity in HCC patients based on clinical observation and stabilization of disease
- The observation of a decrease in hepatitis C viral load is consistent with CF102's known preclinical anti-viral activity
- CF102 treatment appears to hold promise as a novel therapeutic strategy in the treatment of advanced HCC and viral-related liver diseases
- Enrollment in the dose-confirmation phase of this trial is ongoing